No. 368

WBC 2008

July 6–11
Budapest, Hungary

Evaluation of four fluoroquinolone treatment regimens for acute bovine mastitis

Froyman R.[1], Filios S.[1], Brunner B.[2], Torres A.[1], Hellmann K.[2]

[1] Bayer HealthCare, Animal Health Division, 51368 Leverkusen, Germany
[2] Klifovet, 80689 Munich, Germany

Aim of the study

- Acute mastitis constitutes a major cause of economic loss in dairy industry.
- Acute mastitis accompanied by generalised disease is potentially life threatening and requires systemic antibiotic medication in combination with anti-inflammatory products where appropriate.
- This field study aimed to assess four fluoroquinolone (FQ) treatment regimens for acute bovine mastitis with generalised disease.
- The products tested in this trial have a claim for the treatment of acute coliform mastitis. However acute mastitis with generalised disease may also be associated with gram-positive bacteria.
- As medical intervention is urgent for such cases and microbiological results are not available before onset of treatment, all inclusion compliant cases, irrespective of causative bacterium, were enrolled in this trial.

Materials and methods

- Multicenter GCP trial (9 sites, 28 farms)
- 173 cows: 1st to 7th lactation, 88% ≤3rd lactation
- **Inclusion criteria**
 - Acute mastitis
 Local clinical score (LCS) ≥2
 - Generalised disease
 - General condition score (GCS) ≥3
 - Temperature ≥39.6 °C
- Enrolled randomly at day 0 (inclusion and first treatment) until ~25 bacteriologically positive cases were obtained per group.

Study population

	Group 1	Group 2	Group 3	Group 4
	ENR, 5 mg, day 0, [1]	ENR, 5 mg, day 0, 1	DAN, 6 mg, day 0, [2]	MAR, 2 mg, day 0, 1, 2
Number enrolled and treated	**37**	**48**	**43**	**45**
Number enrolled, treated and microbiologically positive	**24**	**31**	**29**	**24**

- **Treatments**
 - All antimicrobial treatments were given intravenously. In all groups, except group 1, the products were used per label.
 - Products used:
 - Baytril 10% (Bayer HealthCare) : enrofloxacin (ENR)
 - Marbocyl 10% (Vétoquinol): marbofloxacin (MAR)
 - Advocid 180 (Pfizer): danofloxacin (DAN)
 - In cows with temperature at ≥ 39.5°C by day 1, supportive intravenous NSAID treatment was given (0.5 mg meloxicam/kg).
 - Treatment groups

Group 1 (n= 37)	5 mg ENR/kg	1 single injection if clinically required, second dose 24 h later
Group 2 (n= 48)	5 mg ENR/kg	2 daily injections
Group 3 (n= 43)	6 mg DAN/kg	1 single injection if clinically required, second dose 48 h later
Group 4 (n= 45)	2 mg MAR/kg	3 daily injections

Clinical cure rates (%)

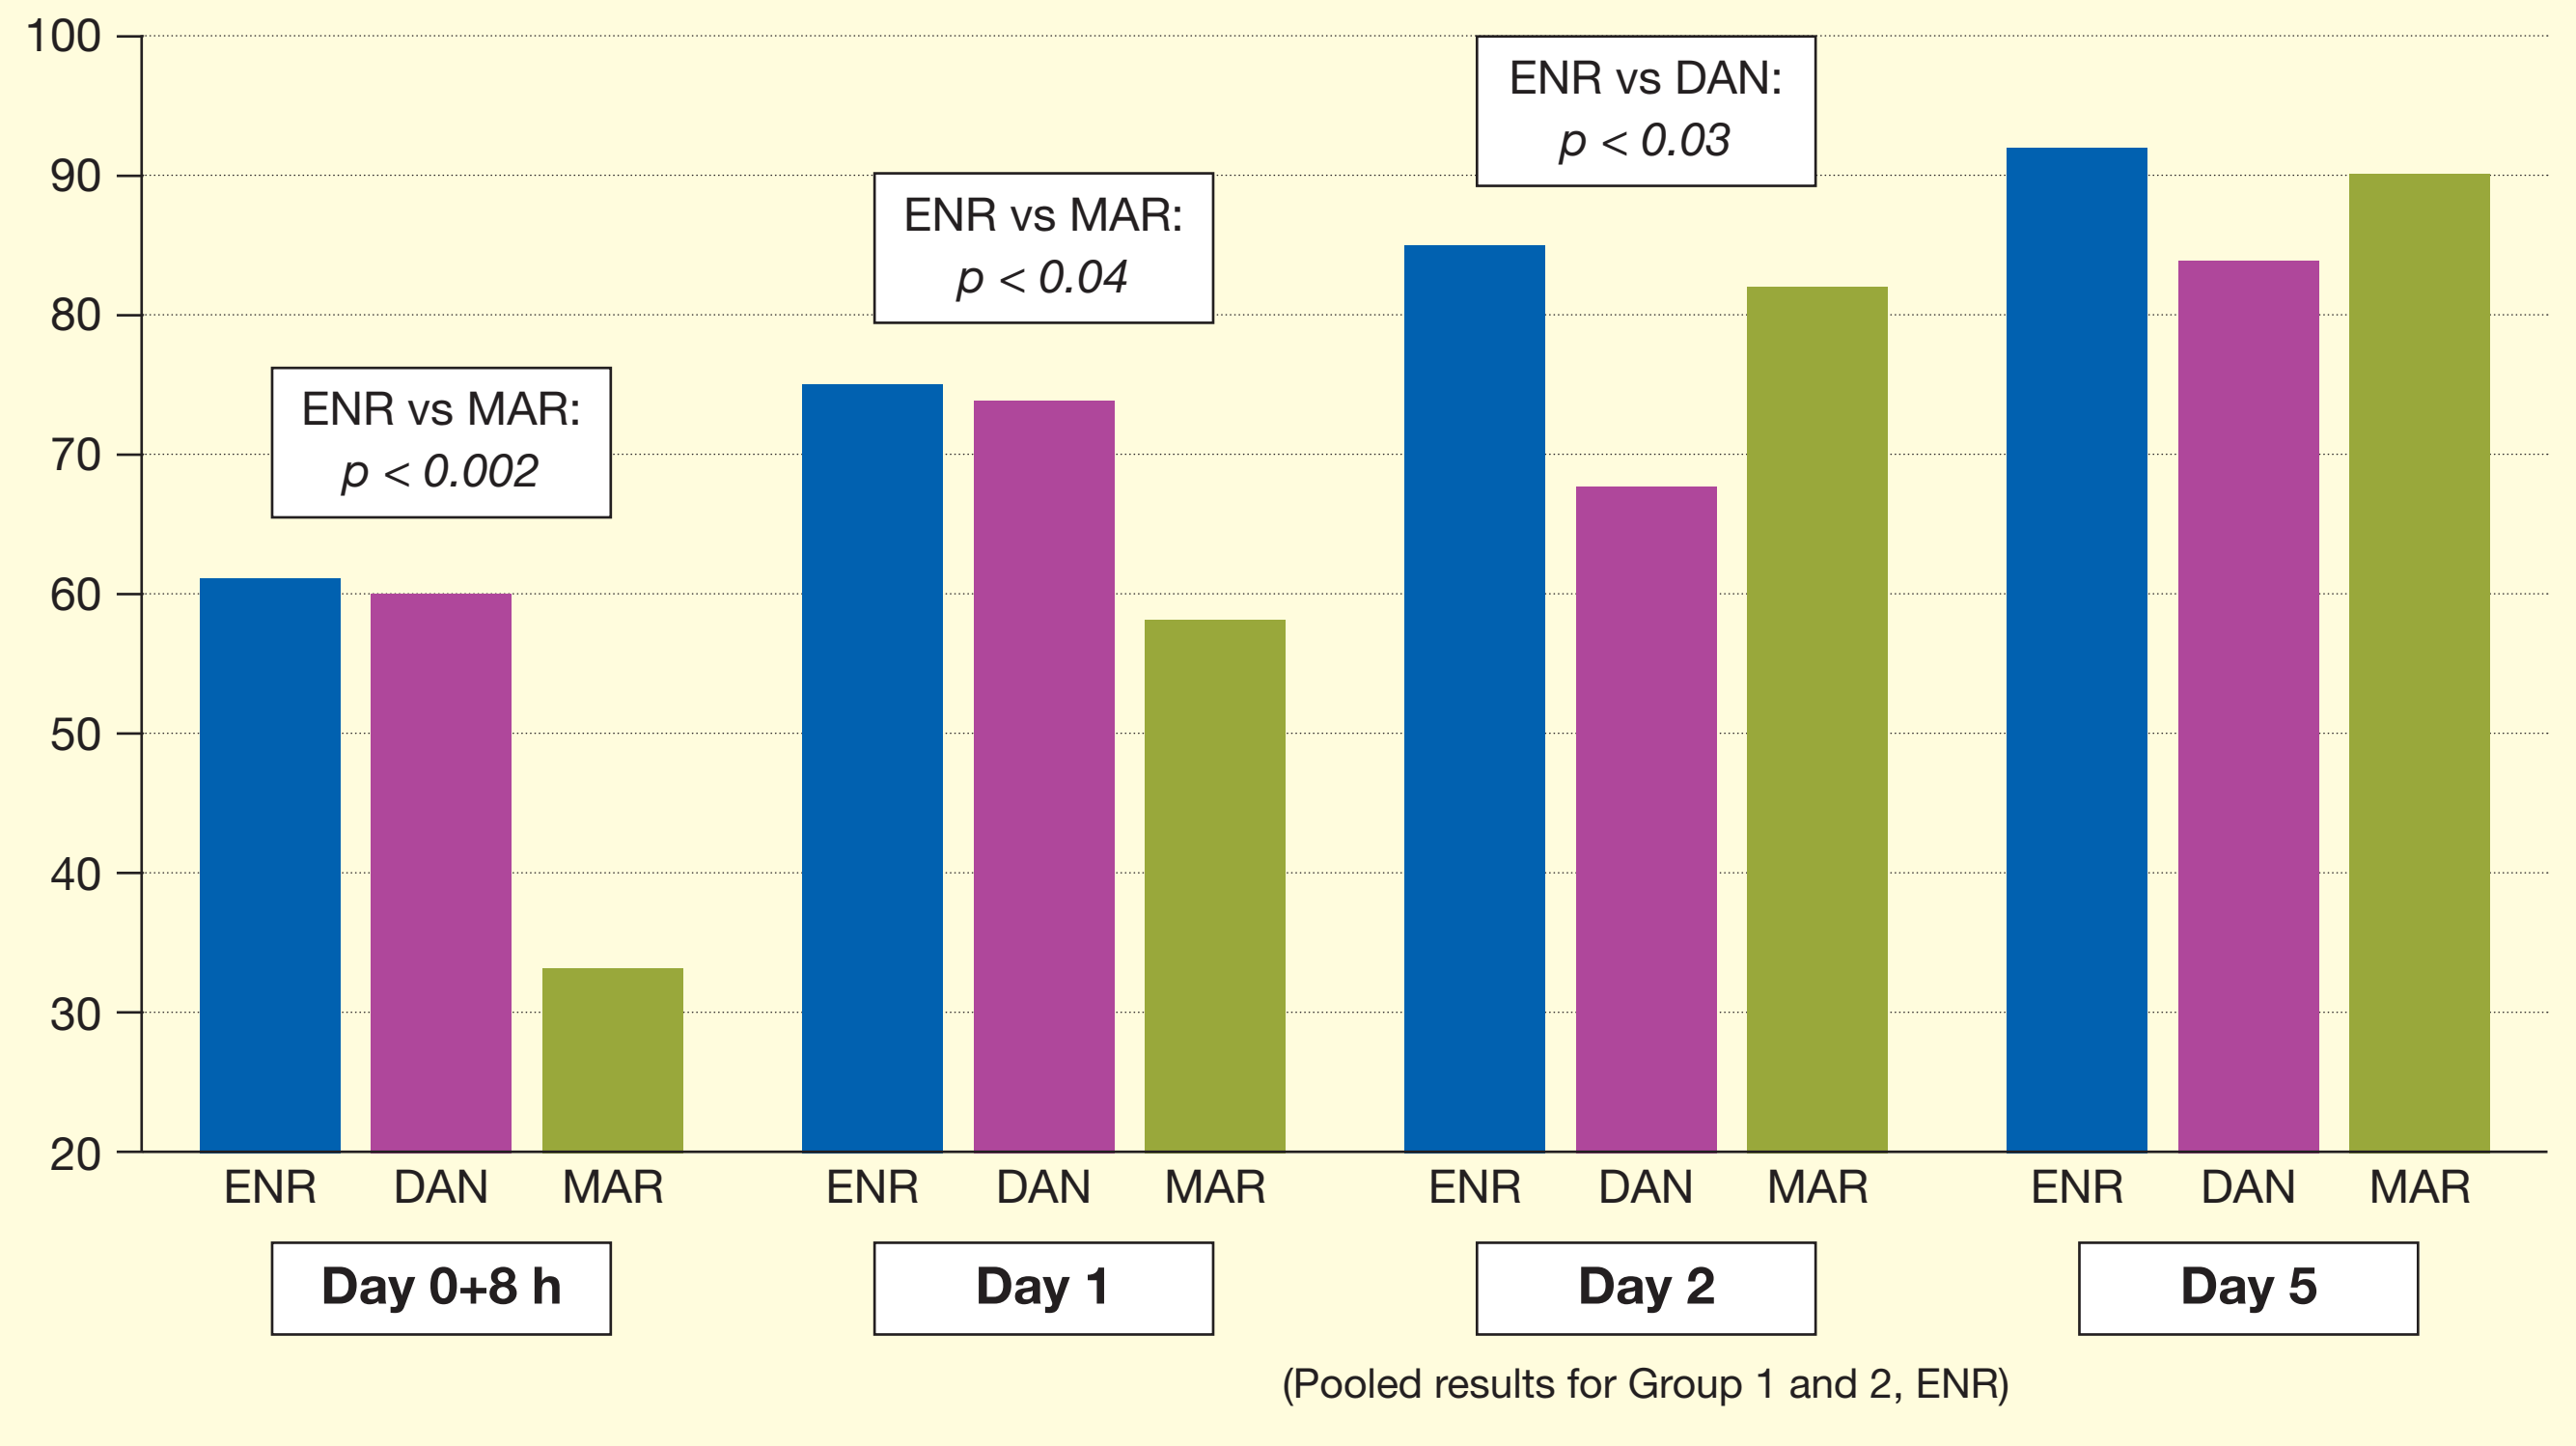
100
90
80
70
60
50
40
30
20
ENR vs MAR: $p < 0.002$
ENR vs MAR: $p < 0.04$
ENR vs DAN: $p < 0.03$
ENR DAN MAR
Day 0+8 h
Day 1
Day 2
Day 5

(Pooled results for Group 1 and 2, ENR)

- **Clinical parameters**
 - Observations
 - Day 0
 - Day 0+8h
 - Day 1, 2, 3, 4 and 5
 - General clinical parameters (rectal temperature, attitude, appetite, pulse rate, rumen movements, respiration rate): general condition score (GCS)
 - Udder inspection (swelling, pain, heat) and milk inspection (flakes/clots, consistency, colour, smell, CMT): local clinical score (LCS)
 - Milk sample for culture at days 0 to 5
 - Statistical analysis. Comparisons between ENR and other FQ treatments
 - ANCOVA for mean scores
 - Chi-square for clinical cure rates (CCR)

Speed of clinical recovery

GCS ◆ down from ≥ 6.5 to < 1.0

LCS ■ down from ≥ 7 to < 3

		Study day					
		0	1	2	3	4	5
Group 1	ENR, 5 mg, day 0, [1]				◆	■	
Group 2	ENR, 5 mg, day 0, 1			◆	■		
Group 3	DAN, 6 mg, day 0, 2						◆■
Group 4	MAR, 2 mg, day 0, 1, 2				◆	■	

- **Efficacy criteria**
 - Clinical cure: decrease of rectal temperature to ≤39.0 C
 - Clinical improvement: decrease of rectal temperature to 39.1-39.5 C
 - Bacteriological cure: negative milk sample at day 5
 - Overall cure rate of clinical mastitis: cases clinically cured and LCS ≤ 1
 - GCS, LCS

Mild yield increase from Day 0 (baseline) to Day 5, as % from baseline

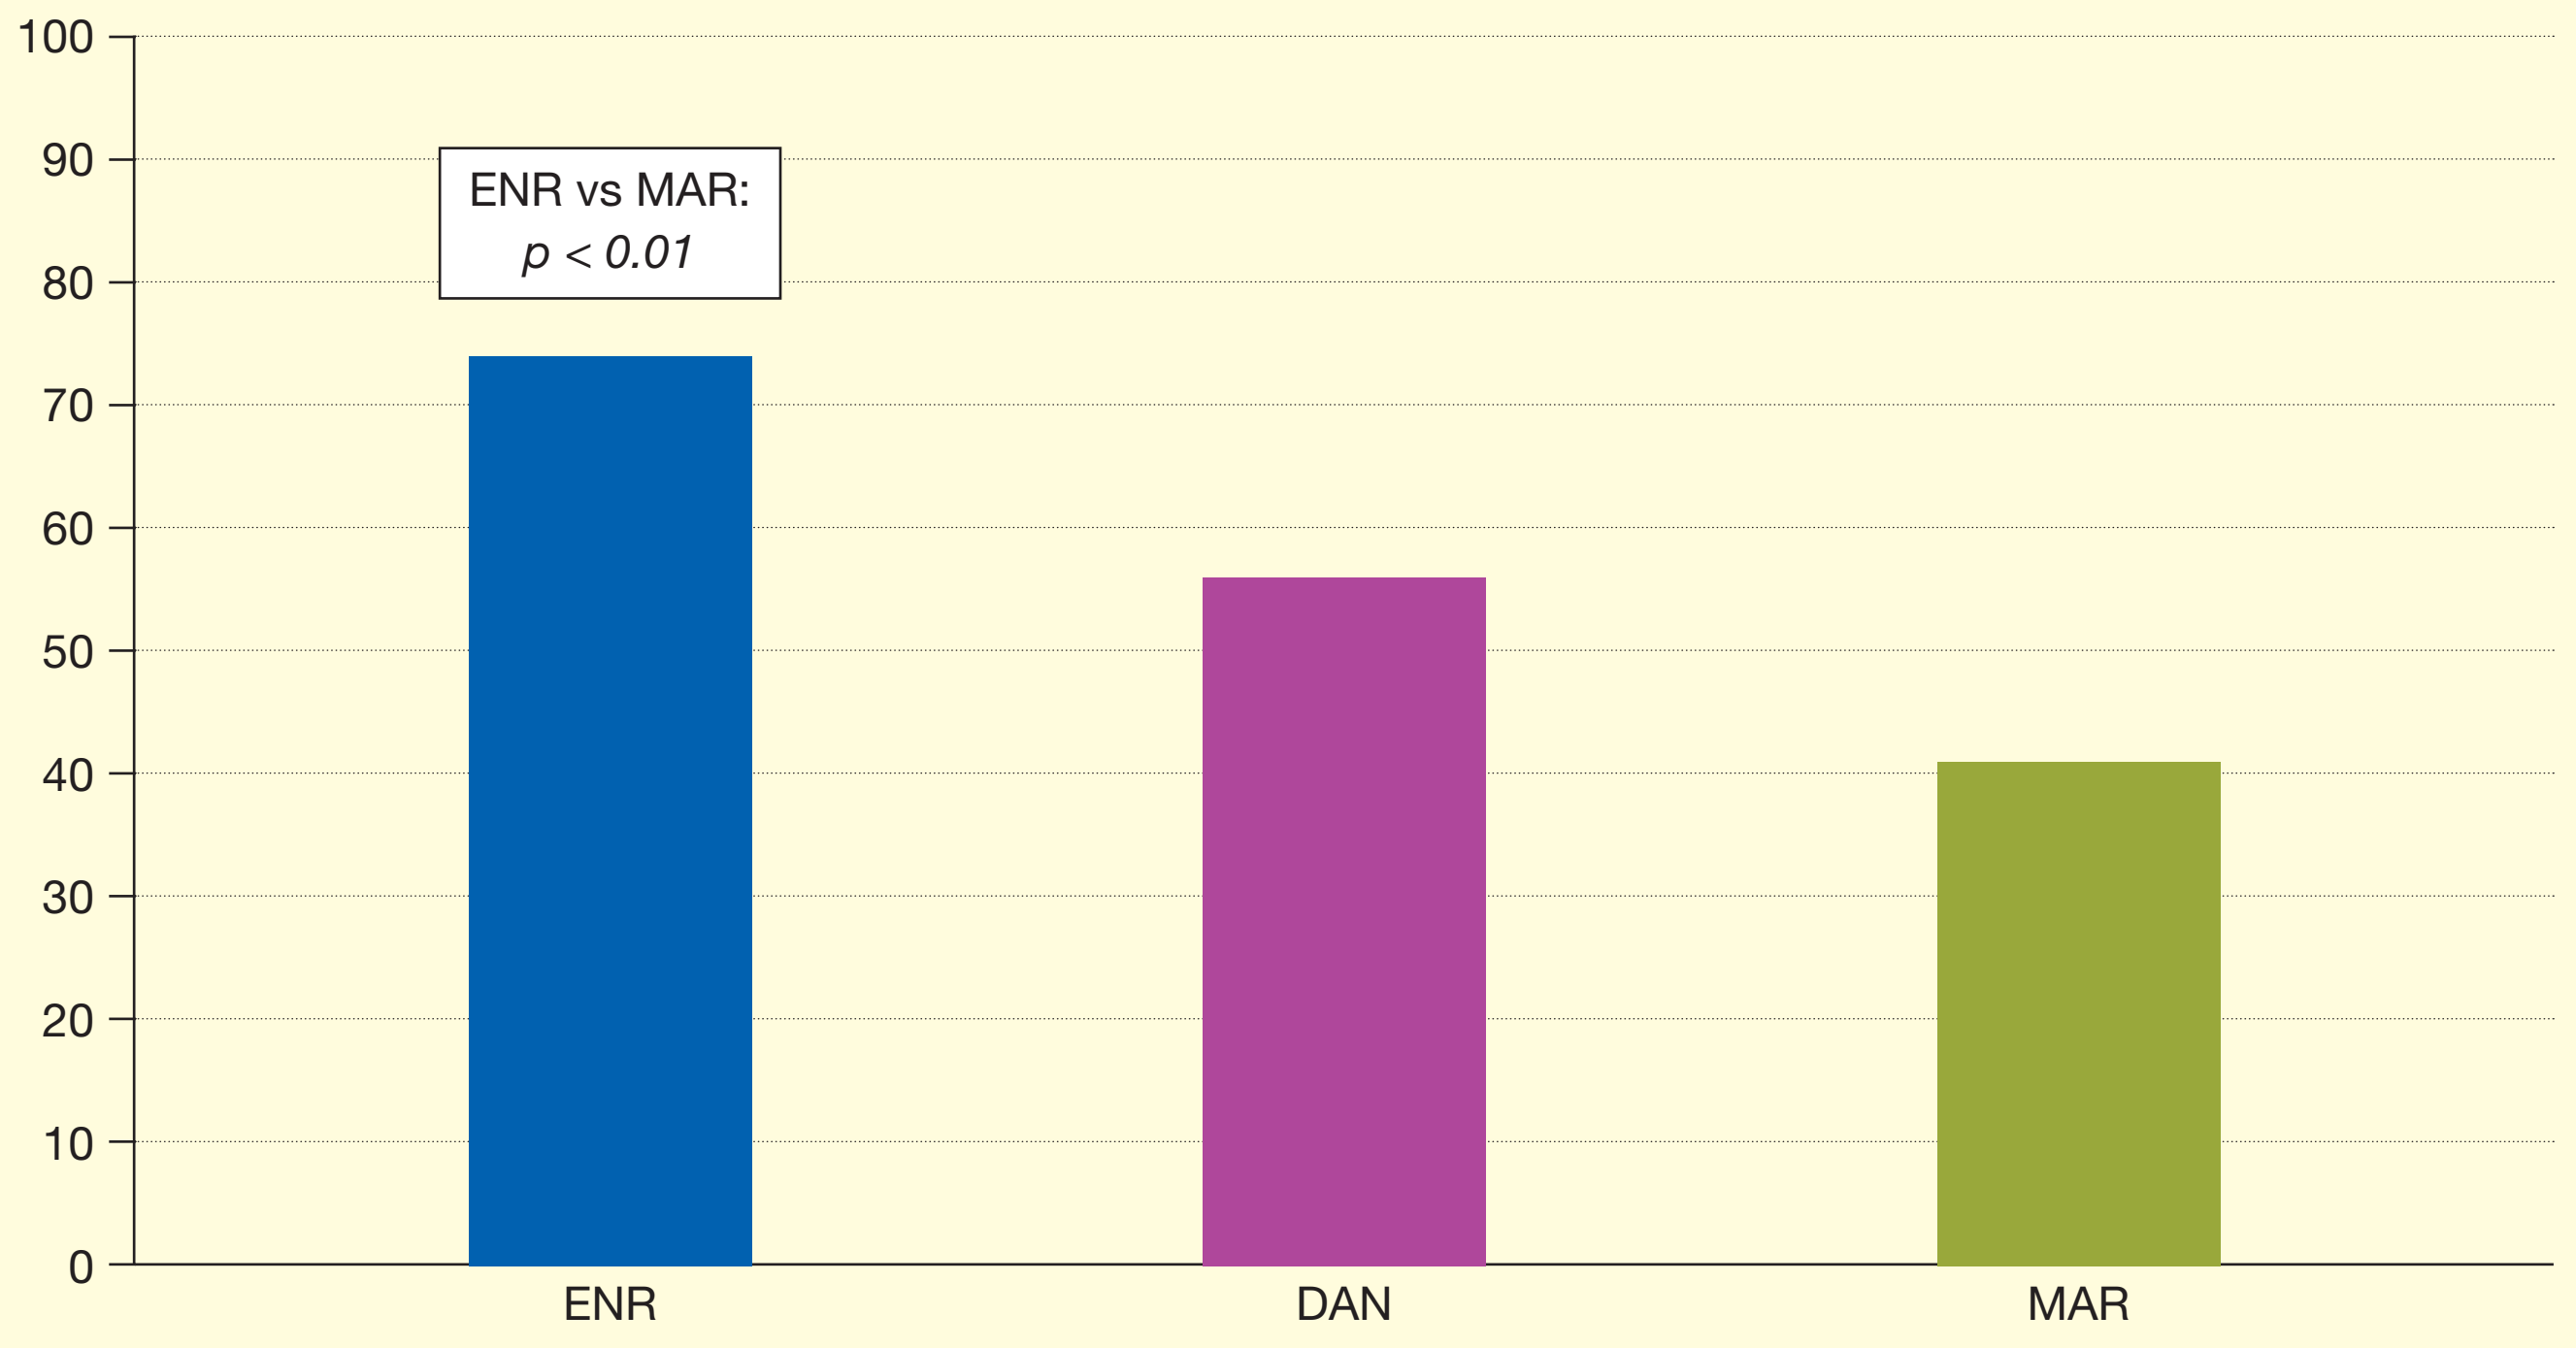
100
90
80
70
60
50
40
30
20
10
0
ENR vs MAR: $p < 0.01$
ENR
DAN
MAR

(Pooled results for Group 1 and 2, ENR)

Results and discussion

- From 173 cows treated, 108 fulfilled all inclusion criteria and were bacteriologically positive, thereof 55% for *E. coli* and 23% for *Str. uberis*.
- Across treatment groups bacteriological cure rate was 79% for coliform mastitis and 54% for *Str. uberis* cases.
- Redosing in group 1 and group 3 was required for 22% and 37% of cows, respectively.
- Fever decreased rapidly in all animals within 8 hours (≥1.5 °C).
- At study end CCRs were 93%, 91%, 84%, and 90% in groups 1, 2, 3 and 4, respectively
- Overall mastitis cure rates (clinically cured and LCS ≤ 1) were 69%, 77%, 58% and 68%, respectively.
- Mean milk yield at day 5 compared to baseline was increased by 74% in ENR, 56% in DAN and 41% in MAR treated cows.
- A dedicated study with a larger population would be needed to substantiate further whether a single intravenous enrofloxacin dose with optional redosing 24 later is as efficacious as 2 daily doses.
- All in all, among FQ treatments, Baytril showed most rapid cure from mastitis symptoms and general disease symptoms and is therefore a preferred therapy for acute mastitis with generalised disease.